ASCO abstract: TPS7092

Trial in progress: First line Treatment of Follicular Lymphoma (FL) with Golcadomide, Rituximab +/- Nivolumab: An Umbrella Randomised Phase 2 Investigator-led Study.

G Chong[1], J Palmer[2], A Barraclough[3], L Churilov[4], C Keane[5], ST Lee[2,6], D Lee[8], S Ratnasingam[9], C Smith[10], EA Hawkes[2,10]

1. Grampians Health, Ballarat, Australia, 2. Olivia Newton-John Cancer Research Institute, Melbourne, Australia, 3. Fiona Stanley Hospital, Perth, Australia., 4. University of Melbourne, Australia. 5. Frazer Institute, University of Queensland, Australia, 6. Dept. Molecular Imaging and Therapy, Austin Health, Melbourne, Australia, 8. Eastern Health, Box Hill Hospital, Australia, 9. Barwon Health, University Hospital, Geelong, Australia, 10. Austin Health, Melbourne, Australia,

Background

Follicular lymphoma (FL) is commonly initially treated with chemotherapy plus anti-CD20 monoclonal antibody, which yields high efficacy but also significant toxicity.[1,2]

FL patients are largely aged >65 and may require treatment multiple times over their disease course thus novel regimens which enhance efficacy and minimise toxicity are **highly desirable**.

FL patient outcomes are influenced by tumour microenvironment composition and manipulation with immunotherapy.

Ikaros and Aiolos, key cell development and homeostasis transcription factors are upregulated in FL. Cereblon-modulating compounds (CELMoDs) such as golcadomide (CC-99282; Figure 1) promote degradation of these with demonstrated efficacy alone or with rituximab in FL (responses-ORR- up to 75% in heavily pre-treated FL pts).[3-5]

We reported favourable efficacy of nivolumab a PD1 inhibitor plus R in 1L treatment of FL (ORR 92% complete response CR 54%).[7-8]

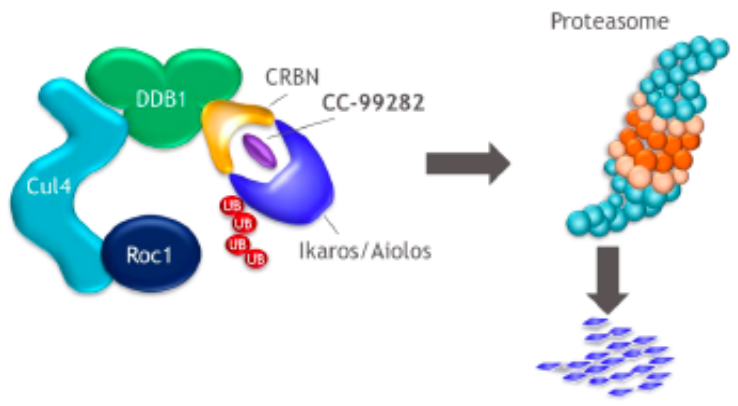
Proteasome
DDB1
CRBN
CC-99282
Cul4
Roc1
UB
Ikaros/Aiolos

Figure 1. Golcadomide co-opts CRBN to induce proteasome mediated degradation of Ikaros and Aiolos[6]

This study ('TOP-FLOR'; NCT05788081) builds on these findings and potential immunomodulatory synergy to explore the efficacy and safety of rituximab + golcadomide, a novel CELMoD +/- nivolumab in treatment-naïve advanced stage FL pts.

Methods

This is an open label multicentre umbrella Bayesian Optimal randomised Phase II trial.

The study will randomise up to 40 eligible pts 1:1 to receive rituximab, golcadomide +/- nivolumab (20 per arm) from 5 Australian sites.

Eligibility includes:

- Patients aged ≥18 years,
- Previously-untreated, stage II-IV, grade 1-3A FL
- Performance status of 0-1
- Requiring systemic therapy.

Planned treatments:

Arm A: 8 cycles (28 days each) of golcadomide 0.4 mg po daily on days 1- 14 plus rituximab 375 mg/m2 IV day 1;
Arm B: Per Arm A plus nivolumab 480mg IV day 1

All patients in partial or complete response at end of induction receive 12-weekly rituximab maintenance (8 doses). Safety assessments will be performed prior to each cycle in accordance with CTCAE V5.0. This is illustrated in Figure 2.

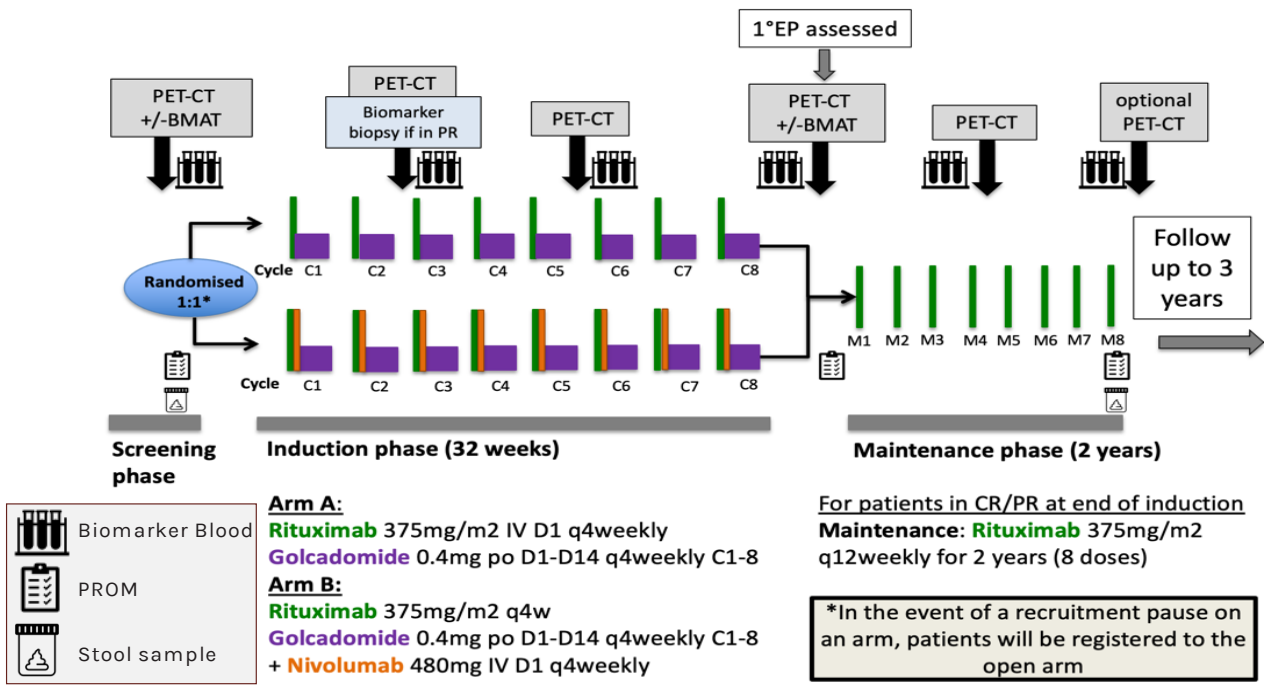
1°EP assessed
PET-CT +/-BMAT
PET-CT
Biomarker biopsy if in PR
PET-CT
PET-CT +/-BMAT
PET-CT
optional PET-CT
Randomised 1:1*
Cycle C1 C2 C3 C4 C5 C6 C7 C8
Cycle C1 C2 C3 C4 C5 C6 C7 C8
M1 M2 M3 M4 M5 M6 M7 M8
Follow up to 3 years
Screening phase
Induction phase (32 weeks)
Maintenance phase (2 years)
Biomarker Blood
PROM
Stool sample
Arm A:
Rituximab 375mg/m2 IV D1 q4weekly
Golcadomide 0.4mg po D1-D14 q4weekly C1-8
Arm B:
Rituximab 375mg/m2 q4w
Golcadomide 0.4mg po D1-D14 q4weekly C1-8
+ Nivolumab 480mg IV D1 q4weekly
For patients in CR/PR at end of induction
Maintenance: Rituximab 375mg/m2 q12weekly for 2 years (8 doses)
*In the event of a recruitment pause on an arm, patients will be registered to the open arm

Figure 2. TOP-FLOR Study treatment and intervention schedule

Funding: This trial is supported by an investigator-sponsored research grant from Bristol-Myers Squibb. Nivolumab and golcadomide supplied by Bristol-Myers Squibb.
Acknowledgements We thank the clinical trials teams at all sites for their work and care of the patients who have participated in the study.

Primary endpoint

CR rate in the absence of prohibitive toxicity in accordance with CTCAE V5.0 after eight cycles of rituximab, golcadomide +/- nivolumab.

Secondary endpoints

- Overall toxicity, response rate (according to modified Lugano criteria)
- Time to treatment failure
- Progression-free survival & overall survival

Exploratory endpoints

- Patient reported outcomes (PRO) as measured by EORTC-QLQ-c30 and FACT-Lym
- PET radiomics
- Tissue, blood and stool immune and genomic biomarkers

Planned Statistical Analysis

Interim assessment of 1^{O} endpoint will be conducted for 11 pts in each arm, arms will be closed for futility if fewer than 7 pts achieve the primary endpoint. Treatment arms will not be formally compared. Survival probabilities will be reported using Kaplan-Meier analysis.

Status

This trial (NCT05788081) received IRB approval on 14 July 2023 at Austin Health, Heidelberg, Australia.

Approval number: HREC/88597/Austin-2023. Trial sponsor: Olivia Newton-John Cancer Research Institute.

Five sites have been activated and are recruiting participants: Austin Health, Barwon Health, Grampians Health, Fiona Stanley Hospital and Eastern Health. First site was activated 31 Aug 2023. 23 participants have been recruited, 22 commenced treatment to date.

References

1. Marcus R, Davies A, Ando K, Klapper W, Opat S, Owen C, et al. Obinutuzumab for the First-Line Treatment of Follicular Lymphoma. *N Engl J Med.* 2017;377(14):1331-1344.
2. Rummel MJ, Niederle N, Maschmeyer G, et al.; Study group indolent Lymphomas (StiL). Bendamustine plus rituximab versus CHOP plus rituximab as first-line treatment for patients with indolent and mantle-cell lymphomas: an open-label, multicentre, randomised, phase 3 non-inferiority trial. Lancet. 2013 Apr 6;381(9873):1203-10. doi: 10.1016/S0140-6736(12)61763-2. Epub 2013 Feb 20. Erratum in: Lancet. 2013 Apr 6;381(9873):1184. PMID: 23433739.
3. Morschhauser F, Nastoupil L, Feugier P, et al: Six-Year Results From RELEVANCE: Lenalidomide Plus Rituximab (R. J Clin Oncol 40:3239-3245, 2022
4. Michot J-M, Chavez JC, Carpio C, et al: Clinical Activity of CC-99282, a Novel, Oral Small Molecule Cereblon E3 Ligase Modulator (CELMoD) Agent, in Patients (Pts) with Relapsed or Refractory Non-Hodgkin Lymphoma (R/R NHL) - First Results from a Phase 1, Open-Label Study. Blood 138:3574-3574, 2021
5. Michot, J.-M. Chavez, J. C. Carpio, C; et.al., S216: Clinical activity of CC-99282, a cereblon E3 ligase modulator (CELMOD) agent, in patients (pts) with relapse/refractory non-Hodgkin lymphoma (R/R NHL) – Results from a phase 1, open-label study. HemaSphere 6():p 117-118, June 2022. | DOI: 10.1097/01.HS9.0000843756.99251.20
6. Michot, J-M. Aggressive lymphoma workshop presentation, Bologna, 2023
7. Barraclough A, Chong G, Gilbertson M, et al: Immune Priming with Single-Agent Nivolumab Followed By Combined Nivolumab & Rituximab Is Safe and Efficacious for First-Line Treatment of Follicular Lymphoma; Interim Analysis of the '1st FLOR' Study. Blood 134:1523-1523, 2019
8. Hawkes EA, Lee ST, Chong G, et al: Immune priming with nivolumab followed by nivolumab and rituximab in first-line treatment of follicular lymphoma: The phase 2 1st FLOR study. Journal of Clinical Oncology 39:7560-7560, 2021
9. Hawkes EA: 1st FLOR: nivolumab and rituximab in follicular lymphoma, International Conference on Malignant Lymphoma, The Video Journal of Hematological Oncology., 2021